NCT05739981

Abstract # TPS5633: A Phase II Study Evaluating the effect of IMNN-001 on Second Look Laparoscopy (SLL) when Administered in Combination with Bevacizumab (BEV) and Neoadjuvant Chemotherapy (NACT) in Patients Newly Diagnosed with Advanced Epithelial Ovarian Cancer (EOC)

Amir A. Jazaeri[1], Rachel N. Grisham[2], Stephanie Gaillard[3], Jeffrey A. How[1], Sebastien Hazard[4], Lauren Musso[4], Chrisann Kyi[2]

[1]University of Texas, MD Anderson Cancer Center; [2]Memorial Sloan Kettering Cancer Center; [3]Johns Hopkins Medicine; [4]Imunon, Inc.

Purpose

IMNN-001 comprises a DNA plasmid encoding an interleukin-12 (IL-12) gene and a synthetic polymer facilitating plasmid delivery. IMNN-001 is designed to be delivered locally, offering the potential for cytokines to be expressed specifically in the tumor micro-environment with the goal of achieving increased efficacy while minimizing potential systemic toxicity. It has been shown to exhibit a favorable benefit/risk ratio when administered intraperitoneally (IP), in combination with intravenous platinum-taxane chemotherapy in women with advanced EOC in earlier Phase I & II studies. The 201-21-202 study (NCT05739981) is a multi-center, randomized, open-label phase II study with safety lead-in which hypothesizes the addition of IMNN-001 to neoadjuvant & adjuvant carboplatin-paclitaxel and bevacizumab (chemo + BEV) reduces the risk of histologically documented minimal residual disease (MRD) as determined by SLL by 50%.

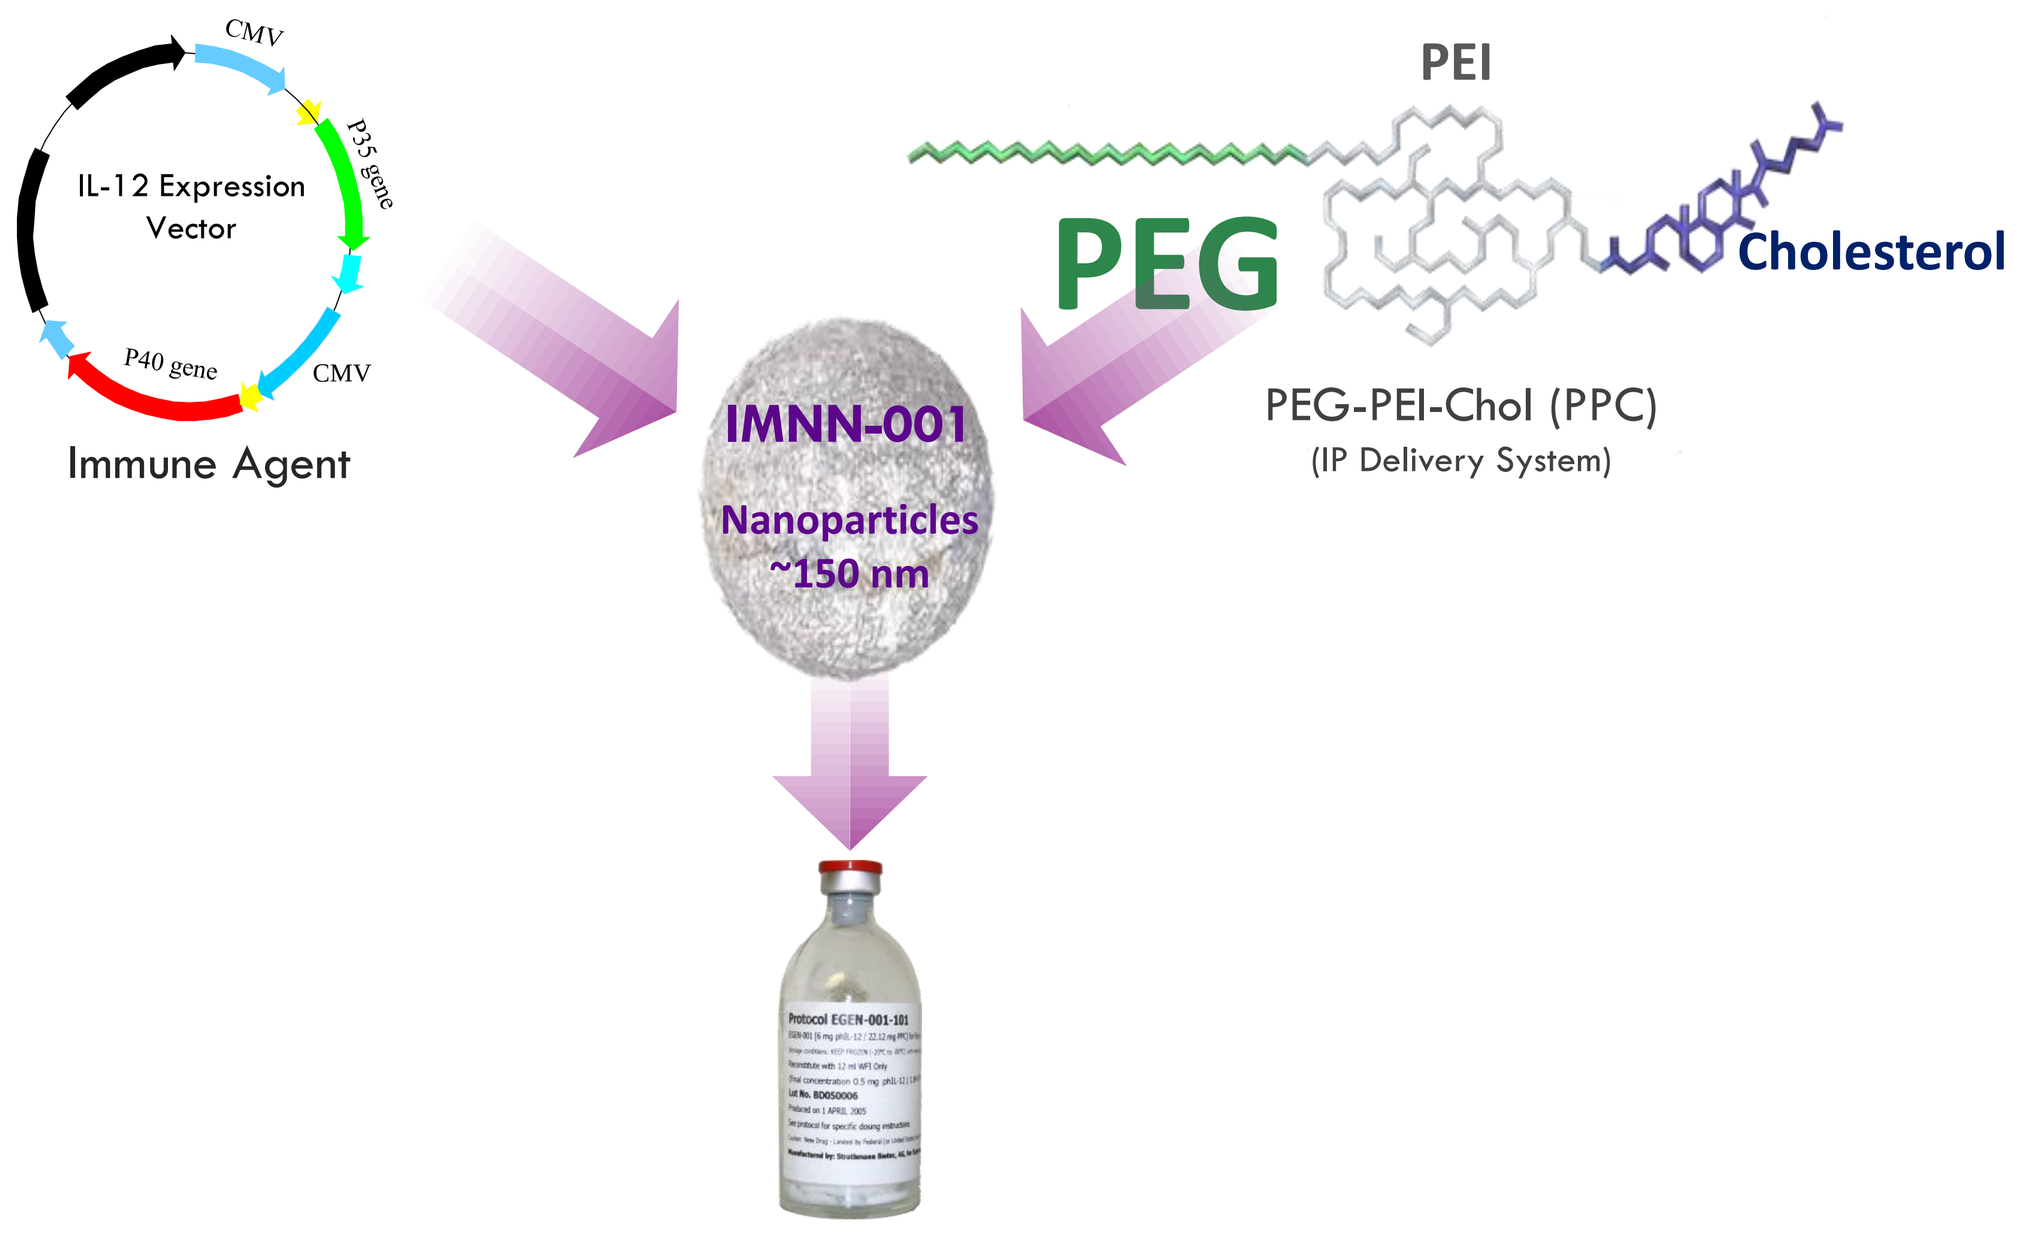
CMV
IL-12 Expression Vector
P35 gene
P40 gene
CMV
Immune Agent
PEI
PEG
Cholesterol
PEG-PEI-Chol (PPC)
(IP Delivery System)
IMNN-001
Nanoparticles ~150 nm

IMNN-001

Intraperitoneal (IP) Infusion Produces Durable Local Expression of IL-12 in the Peritoneal Cavity (PC)

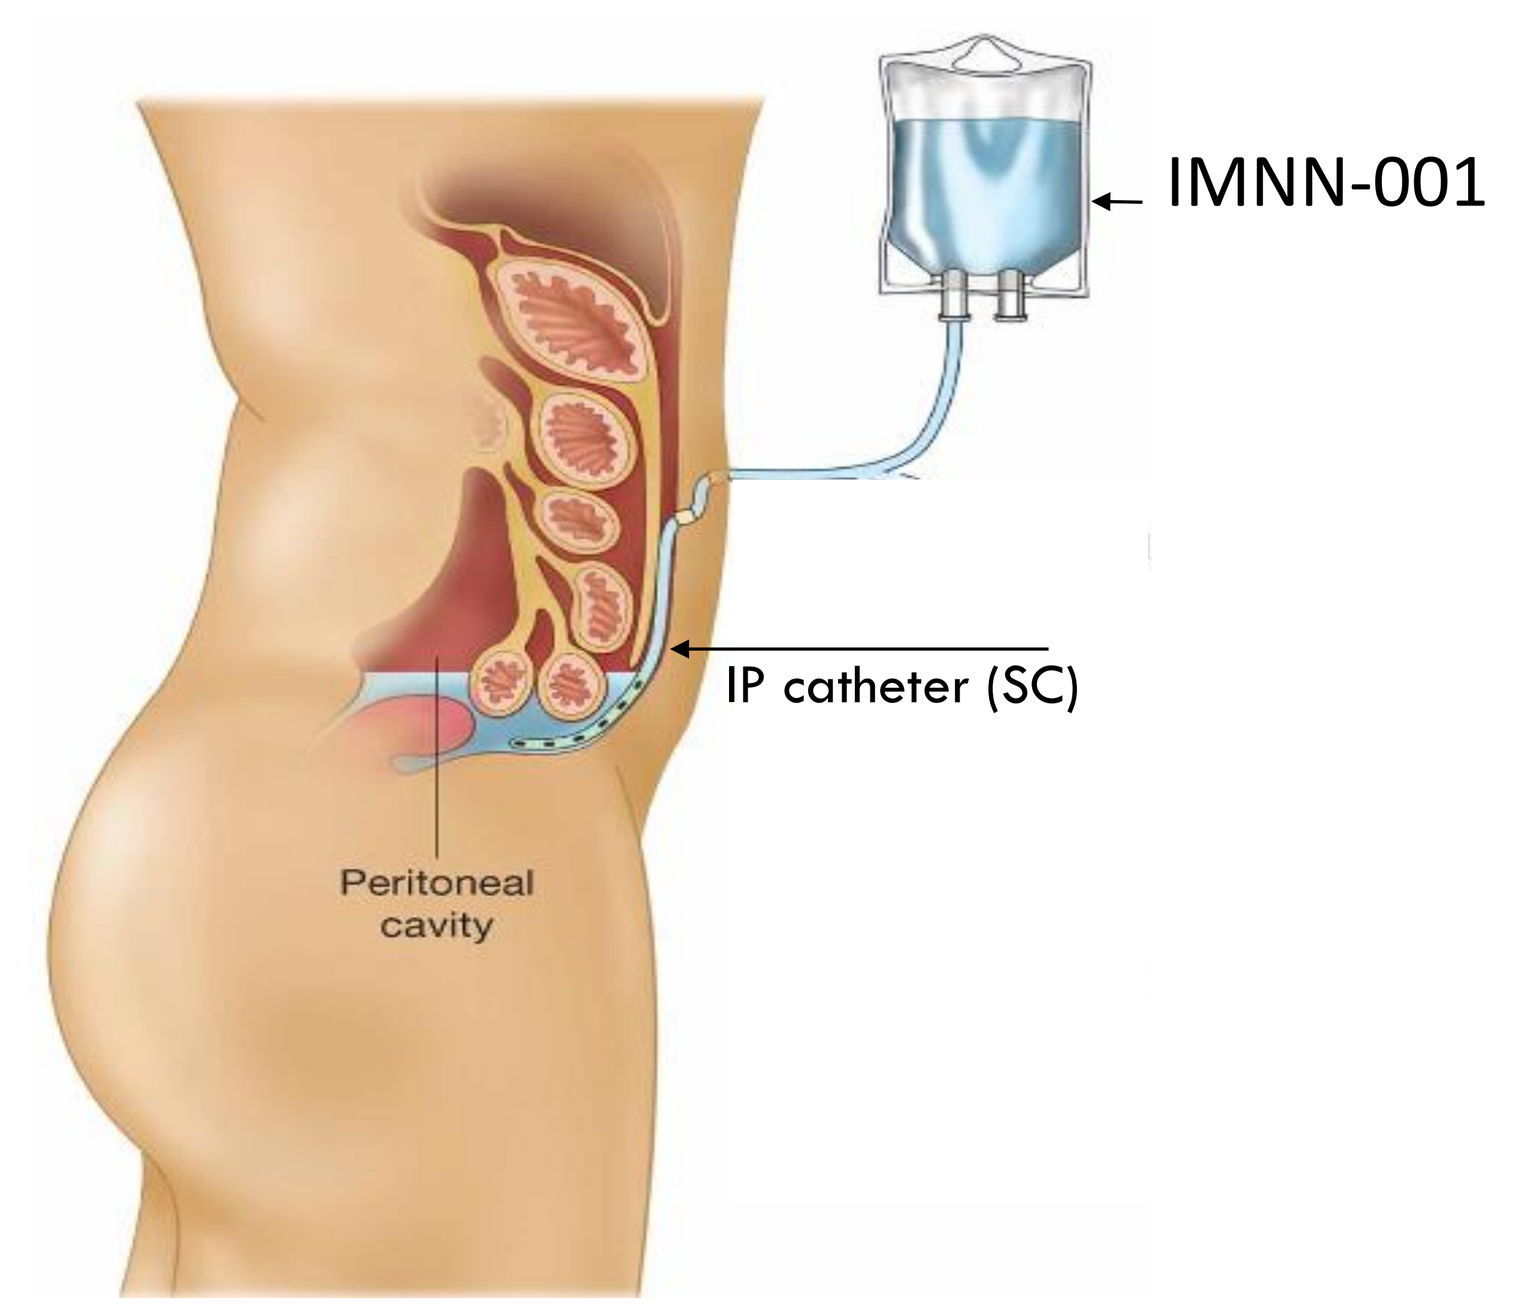
IMNN-001
IP catheter (SC)
Peritoneal cavity

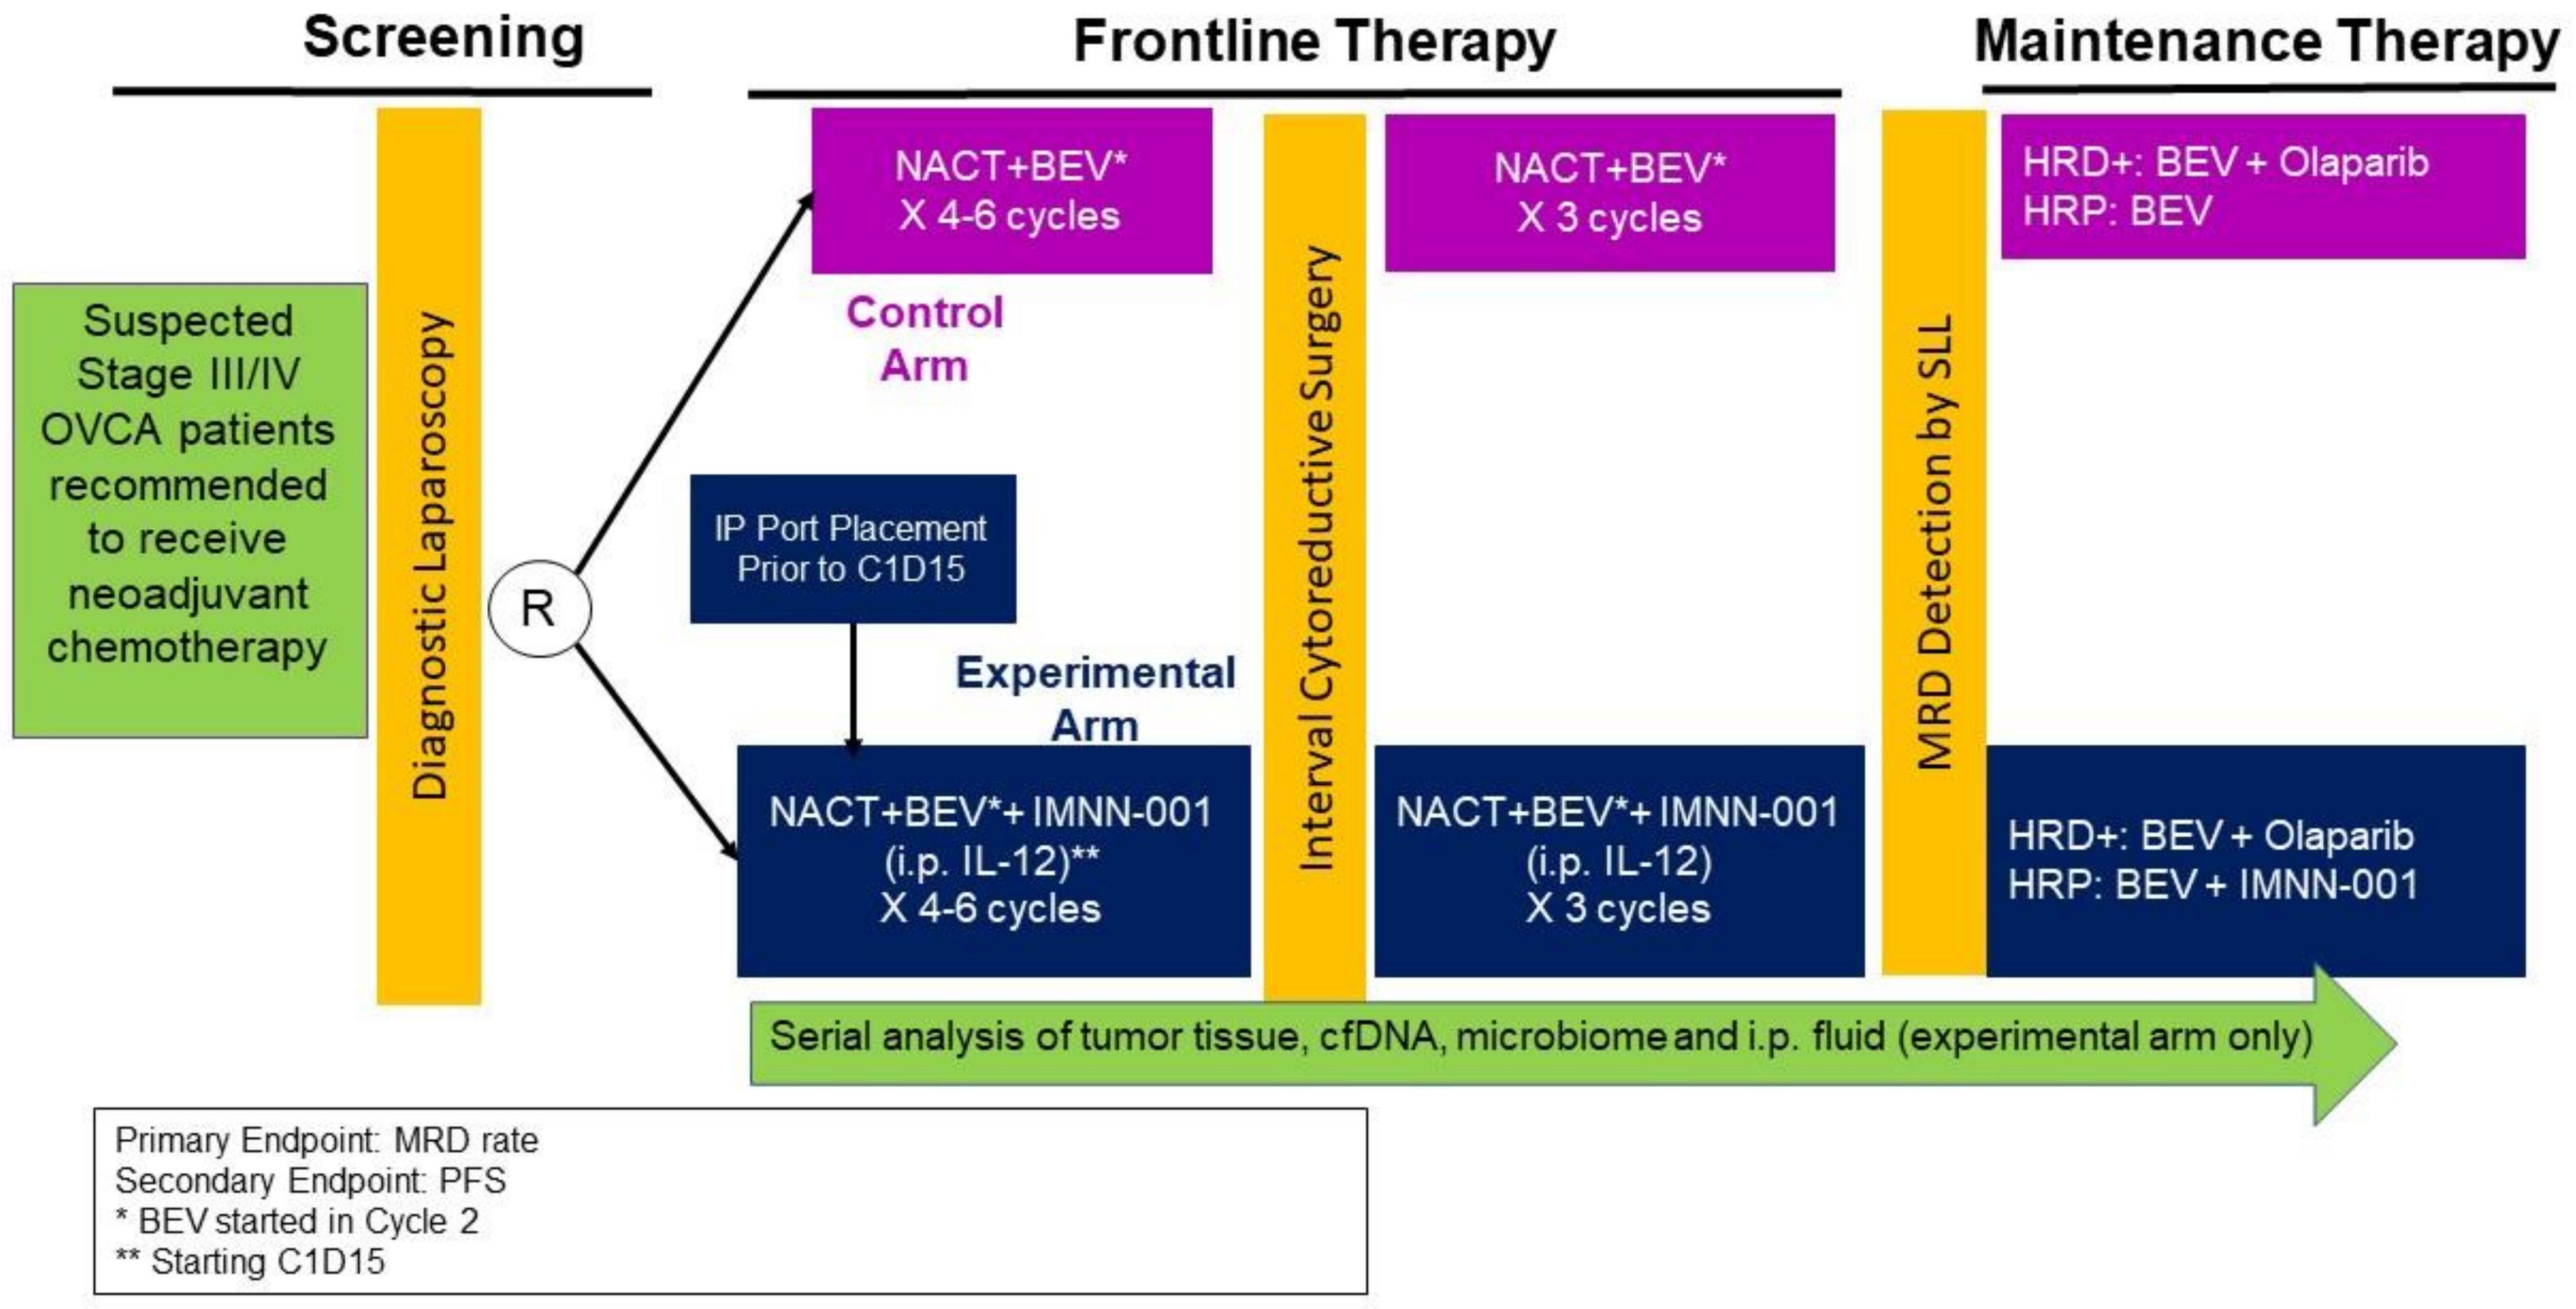
Screening
Frontline Therapy
Maintenance Therapy
Suspected Stage III/IV OVCA patients recommended to receive neoadjuvant chemotherapy
Diagnostic Laparoscopy
R
NACT+BEV* X 4-6 cycles
Control Arm
IP Port Placement Prior to C1D15
Experimental Arm
NACT+BEV*+ IMNN-001 (i.p. IL-12)** X 4-6 cycles
Interval Cytoreductive Surgery
NACT+BEV* X 3 cycles
NACT+BEV*+ IMNN-001 (i.p. IL-12) X 3 cycles
MRD Detection by SLL
HRD+: BEV + Olaparib
HRP: BEV
HRD+: BEV + Olaparib
HRP: BEV + IMNN-001
Serial analysis of tumor tissue, cfDNA, microbiome and i.p. fluid (experimental arm only)
Primary Endpoint: MRD rate
Secondary Endpoint: PFS
* BEV started in Cycle 2
** Starting C1D15

Translational

All subjects will undergo biospecimen collection and organoid preparation prior to treatment, at the time of ICS and SLL for comprehensive longitudinal genomic, circulating tumor DNA, immune, and microbiome profiling. Subjects in the experimental arm will also undergo peritoneal lavage and collection of fluid and cells for assessment of the peritoneal tumor environment.

Patients and Methods

Patients with newly diagnosed stage III and IV high grade serous EOC who are candidates for NACT will be enrolled into the trial. Initial safety lead-in will evaluate at least 6 patients randomized to the experimental arm. Upon confirmation of safety by the DSMB, a target of ~50 patients will be randomized 1:1 to receive either chemo + BEV with or without IMNN-001. Patients receive at least 4 cycles of NACT followed by interval cytoreductive surgery (ICS), and 3 additional adjuvant cycles of study treatments will be administered. BEV will be included with each cycle except cycle 1, the last cycle of NACT immediately preceding ICS, and the first cycle of adjuvant chemotherapy. In the experimental arm, IMNN-001 will be administered on C1D15 and continue weekly through the last cycle of adjuvant therapy. Following adjuvant therapy, all patients will undergo SLL with peritoneal washing and multiple biopsies. Pathologic detection of residual tumor in any biopsies or peritoneal cytology constitutes presence of MRD. All patients will receive maintenance therapy with Olaparib + BEV, IMNN-001 + BEV, or BEV alone based on their homologous recombination deficiency status and treatment randomization.

MRD STUDY

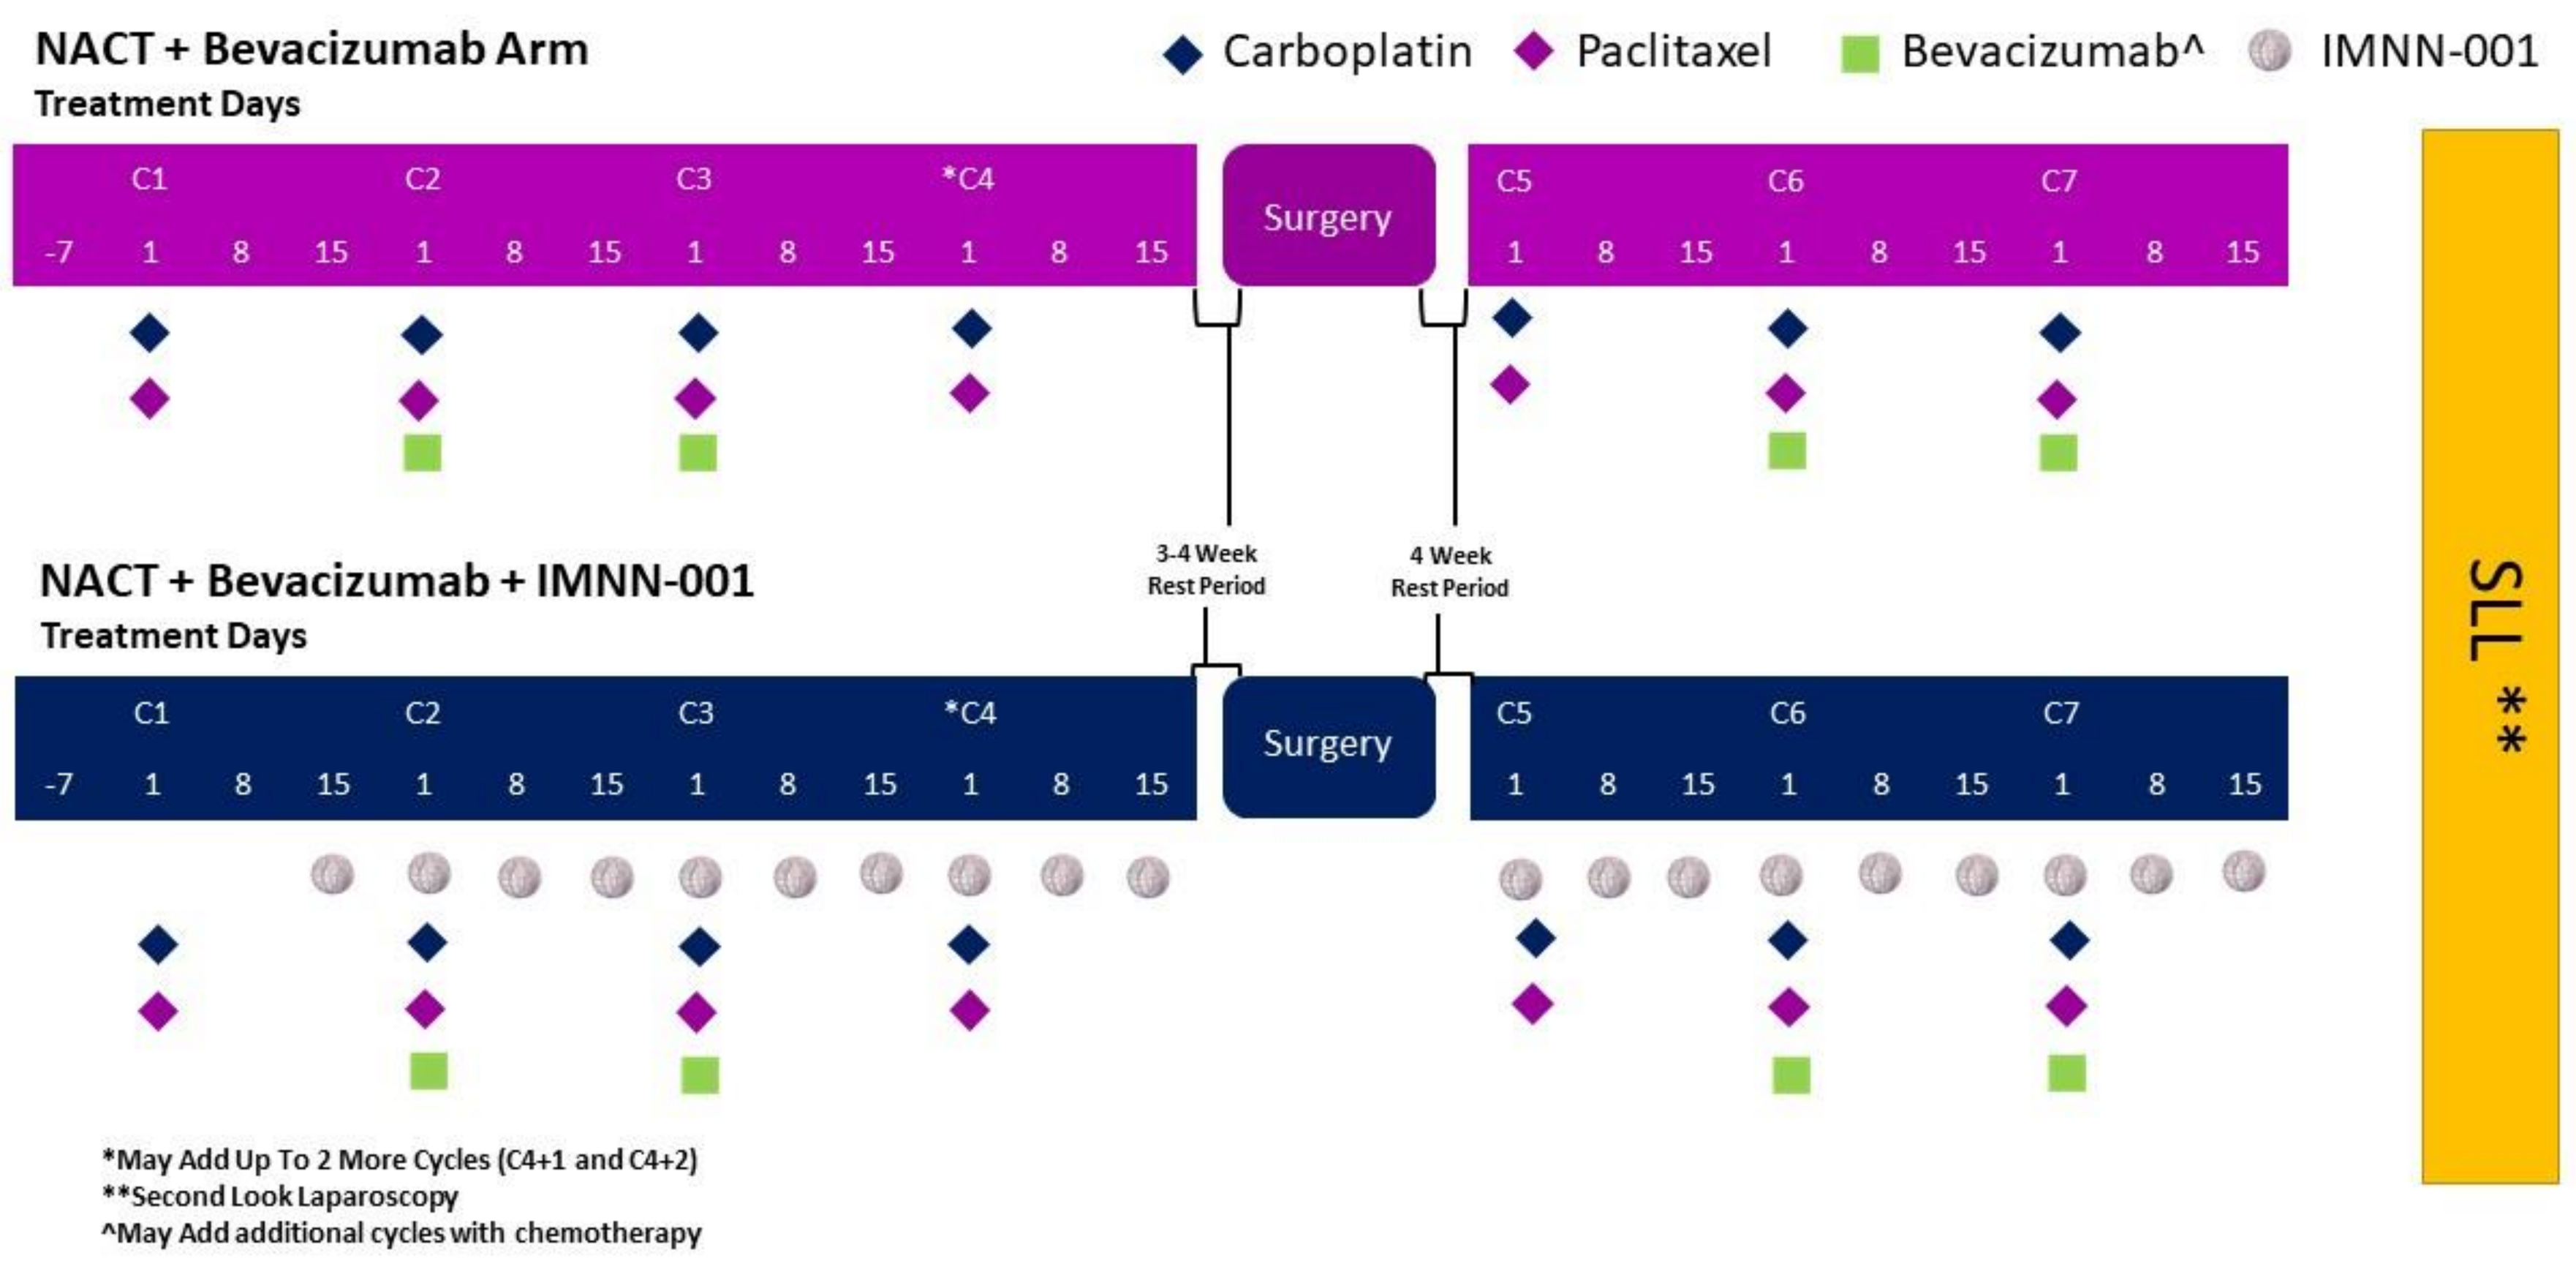
NACT + Bevacizumab Arm
Treatment Days
Carboplatin
Paclitaxel
Bevacizumab^
IMNN-001
C1 C2 C3 *C4
-7 1 8 15 1 8 15 1 8 15 1 8 15
Surgery
C5 C6 C7
1 8 15 1 8 15 1 8 15
3-4 Week Rest Period
4 Week Rest Period
NACT + Bevacizumab + IMNN-001
Treatment Days
SLL **
*May Add Up To 2 More Cycles (C4+1 and C4+2)
**Second Look Laparoscopy
^May Add additional cycles with chemotherapy

Status

The study is actively enrolling and is in the safety lead-in portion.

Endpoints

The primary endpoint is the rate of MRD at SLL. Secondary endpoints include progression-free survival, overall survival, objective response rate, chemotherapy response score, surgical response, and serologic response. All patients will be evaluated for safety.

Contact / Collaborators

For additional information, visit Imunon.com or contact Amir Jazaeri, MD at AAJazaeri@mdanderson.org.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.